Poster P5-16-07

Assessment of Sacituzumab Govitecan in Black Patients From the Phase 3 ASCENT Study in Metastatic Triple-Negative Breast Cancer

Lisa A. Carey,[1] Amelia Zelnak,[2] Hope S. Rugo,[3] Florence Dalenc,[4] Rita Nanda,[5] Michael Danso,[6] Mahasti Saghatchian,[7] Kevin Kalinsky,[8] Nelly Firmin,[9] Manuel Ruiz-Borrego,[10] Anne Favret,[11] Jun Sun,[12] Lee Schwartzberg,[13] Christie Hilton,[14] Coral Omene,[15] Robyn Young,[16] Sara A. Hurvitz,[17] Eliza Harting,[18] See Phan,[18] Aditya Bardia[19]

[1]University of North Carolina Lineberger Comprehensive Cancer Center, Chapel Hill, NC, USA; [2]Northside Hospital, Atlanta, GA, USA; [3]University of California San Francisco Comprehensive Cancer Center, San Francisco, CA, USA; [4]Institut Claudius Regaud, Toulouse, France; [5]The University of Chicago Medical Center, Chicago, IL, USA; [6]Virginia Oncology Associates, PC, Norfolk, VA, USA; [7]Institut Gustave Roussy, Villejuif, France; [8]Winship Cancer Institute, Emory University, Atlanta, GA, USA; [9]Institut Régional du Cancer de Montpellier, Montpellier, France; [10]Hospital Universitario Virgen del Rocío, Sevilla, Spain; [11]Virginia Cancer Specialists, PC, Fairfax, VA, USA; [12]Maryland Oncology Hematology – Clinton Office, Clinton, MD, USA; [13]West Cancer Center, Memphis, TN, USA; [14]Allegheny Health Network Cancer Institute, Pittsburgh, PA, USA; [15]Rutgers Cancer Institute of New Jersey, New Brunswick, NJ, USA; [16]The Center for Cancer and Blood Disorders, Fort Worth, TX, USA; [17]Department of Medicine, Division of Hematology/Oncology, David Geffen School of Medicine, University of California, Los Angeles, Jonsson Comprehensive Cancer Center, Los Angeles, CA, USA; [18]Gilead Sciences Inc., Foster City, CA, USA; [19]Department of Hematology/Oncology, Massachusetts General Hospital Cancer Center, Harvard Medical School, Boston, MA, USA

ASCENT Clinical Trial

Background

Metastatic Triple-Negative Breast Cancer (mTNBC)

- mTNBC is a heterogenous disease with few treatment options and poor outcomes[1-3]
- Single-agent chemotherapy remains standard for previously treated mTNBC, but is associated with low response rates and short progression-free survival (PFS)[4-7]
- TNBC is highly prevalent in Black women,[8] with incidence rates twice as high compared with rates in White women[9,10]
- Black women with TNBC may experience worse clinical outcomes compared with non-Hispanic, White women with TNBC, including a higher risk of mortality potentially due to:[8-9]
 - Disparities (eg, access to health care, delays in treatment, income)
 - Comorbidities
 - Differences in TNBC biology
- As a result, Black patients with TNBC may benefit from novel therapies to improve outcomes

Sacituzumab Govitecan (SG)

- Trop-2 is expressed in all subtypes of breast cancer and linked to poor prognosis[11,12]
- SG is an antibody-drug conjugate (ADC) composed of an anti–Trop-2 antibody coupled to the cytotoxic SN-38 payload via a proprietary, hydrolyzable linker[13]
- SG is distinct from other ADCs (**Figure 1**)[13-17]
 - Antibody highly specific for Trop-2
 - High drug-to-antibody ratio (7.6:1)
 - Internalization and enzymatic cleavage by tumor cell not required for SN-38 (active metabolite of irinotecan) liberation from antibody
 - Hydrolysis of the linker releases SN-38 extracellularly in the tumor microenvironment (bystander effect)
- The U.S. Food and Drug Administration (FDA) granted approval for SG for patients with mTNBC who received ≥2 prior chemotherapies (at least 1 in the metastatic setting) and FDA accelerated approval for patients with metastatic urothelial cancer who received platinum-containing chemotherapy and a checkpoint inhibitor[16,18]
- Results from the confirmatory phase 3 ASCENT study (NCT02574455) demonstrated a significant survival improvement of SG over single-agent chemotherapy, with a manageable safety profile in the second-line or greater (2L+) mTNBC setting[17]
 - Median PFS of 4.8 vs 1.7 months (HR, 0.43; 95% CI, 0.35-0.54) in the full trial population
 - Median overall survival (OS) of 11.8 vs 6.9 months (HR, 0.51; 95% CI, 0.41-0.62) in the full trial population
- A prespecified subgroup analysis was performed to better understand the clinical outcomes of Black patients in the ASCENT study

Figure 1. Sacituzumab Govitecan Antibody-Drug Conjugate

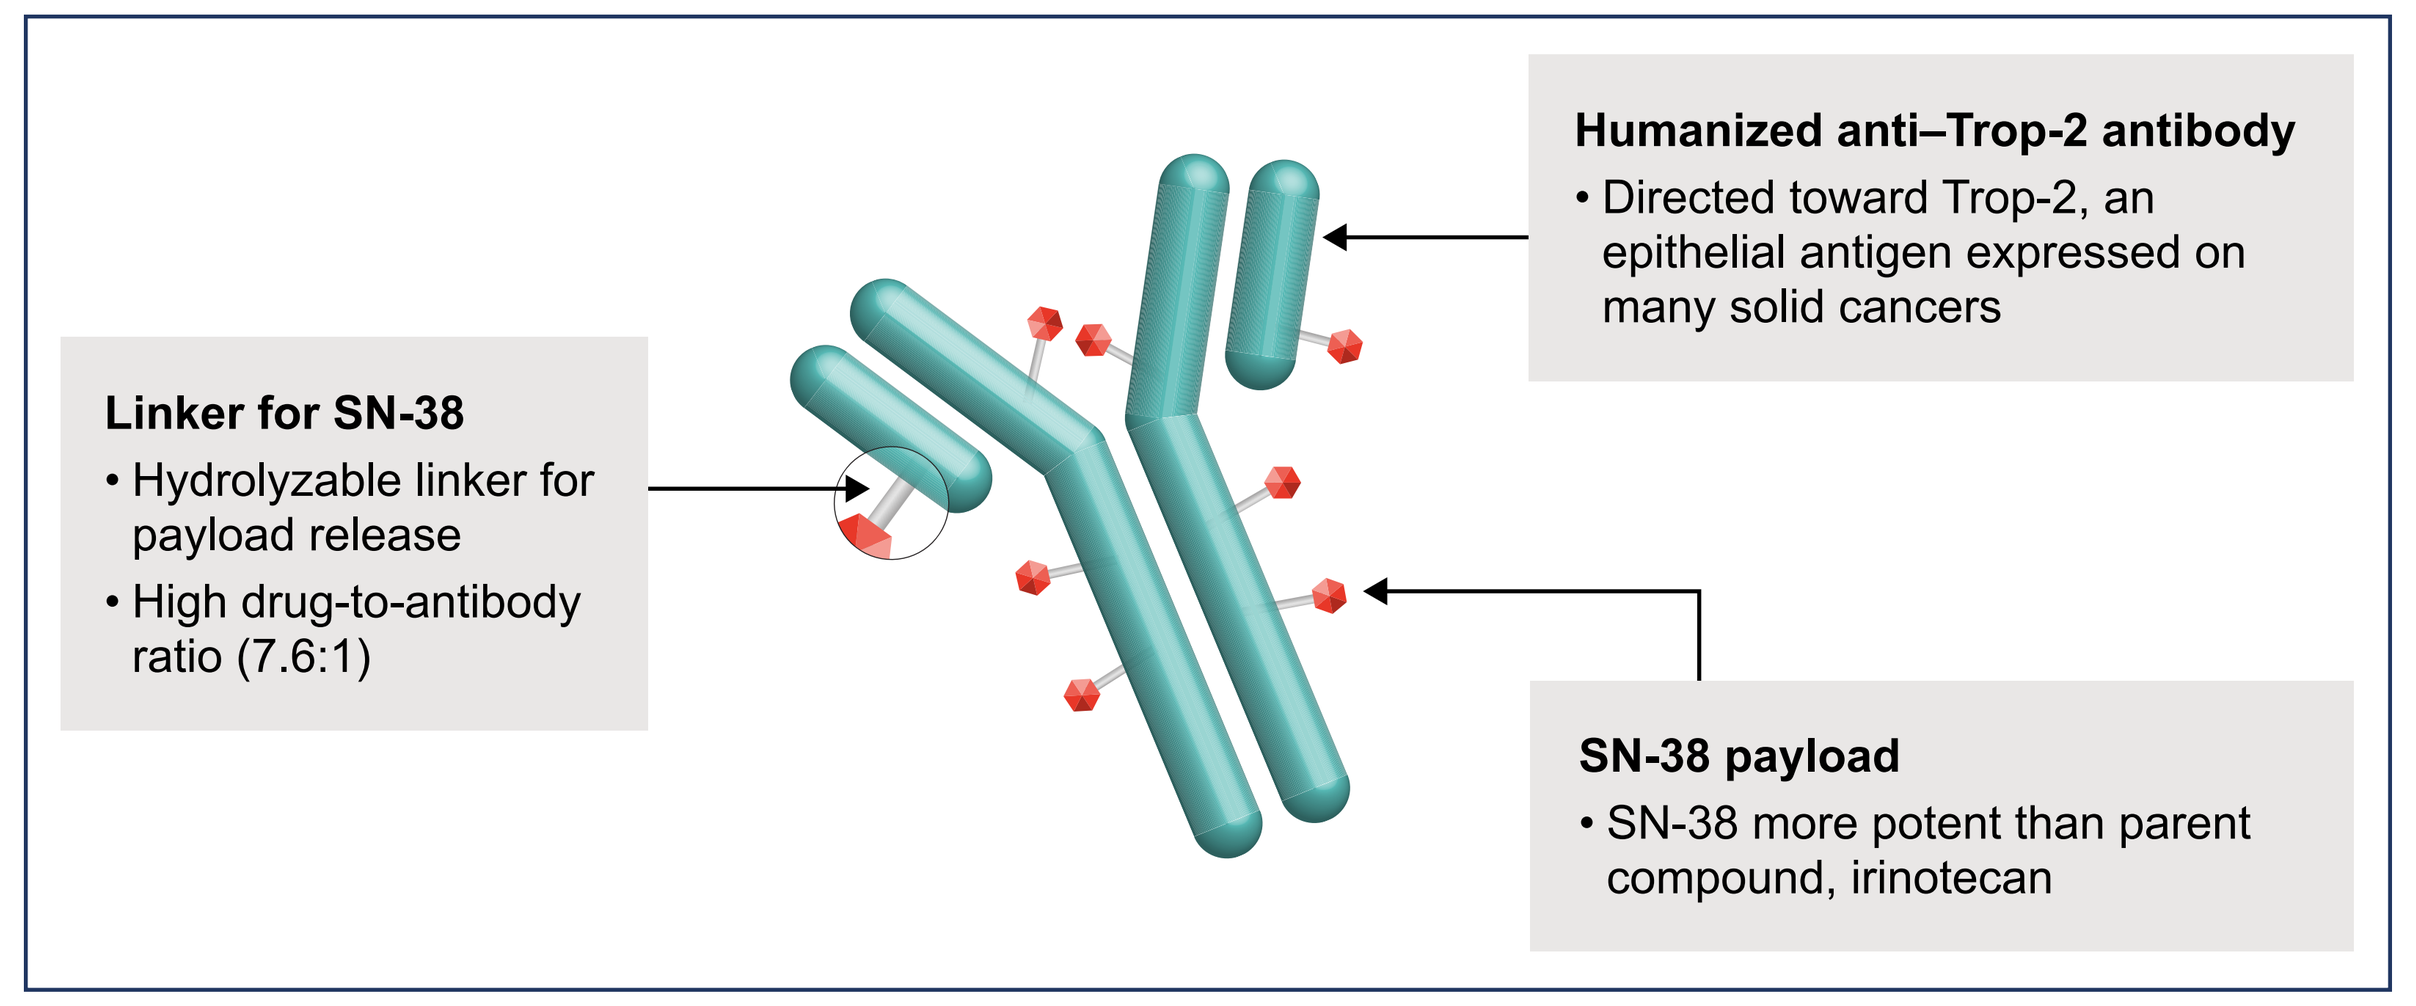

Humanized anti–Trop-2 antibody
• Directed toward Trop-2, an epithelial antigen expressed on many solid cancers
Linker for SN-38
• Hydrolyzable linker for payload release
• High drug-to-antibody ratio (7.6:1)
SN-38 payload
• SN-38 more potent than parent compound, irinotecan

Trop-2, trophoblast cell surface antigen 2.

Methods

- In the phase 3 ASCENT trial, patients with mTNBC refractory to or relapsing after ≥2 prior chemotherapies (at least 1 in the metastatic setting) were randomized 1:1 to receive SG (10 mg/kg IV on days 1 and 8, every 21 days) or single-agent chemotherapy of physician's choice (TPC; capecitabine, eribulin, vinorelbine, or gemcitabine; **Figure 2**)[17]
- Primary endpoint was PFS per RECIST 1.1 by independent review in brain metastases-negative patients; key secondary endpoints included OS, objective response rate (ORR), clinical benefit rate (CBR), and safety
 - Median PFS and ORR were assessed by blind independent central review (BICR) per RECIST 1.1
- In this *post hoc* subgroup analysis, efficacy outcomes were assessed in the full trial population of patients self-reporting Black race who were randomized to receive SG or TPC, including patients with and without known brain metastases
- Safety was analyzed in patients who received ≥1 dose of study drug in this subgroup
- Data cutoff was March 11, 2020

Methods *(continued)*

Figure 2. ASCENT: A Phase 3 Confirmatory Study of SG in Refractory/Relapsed mTNBC (NCT02574455)

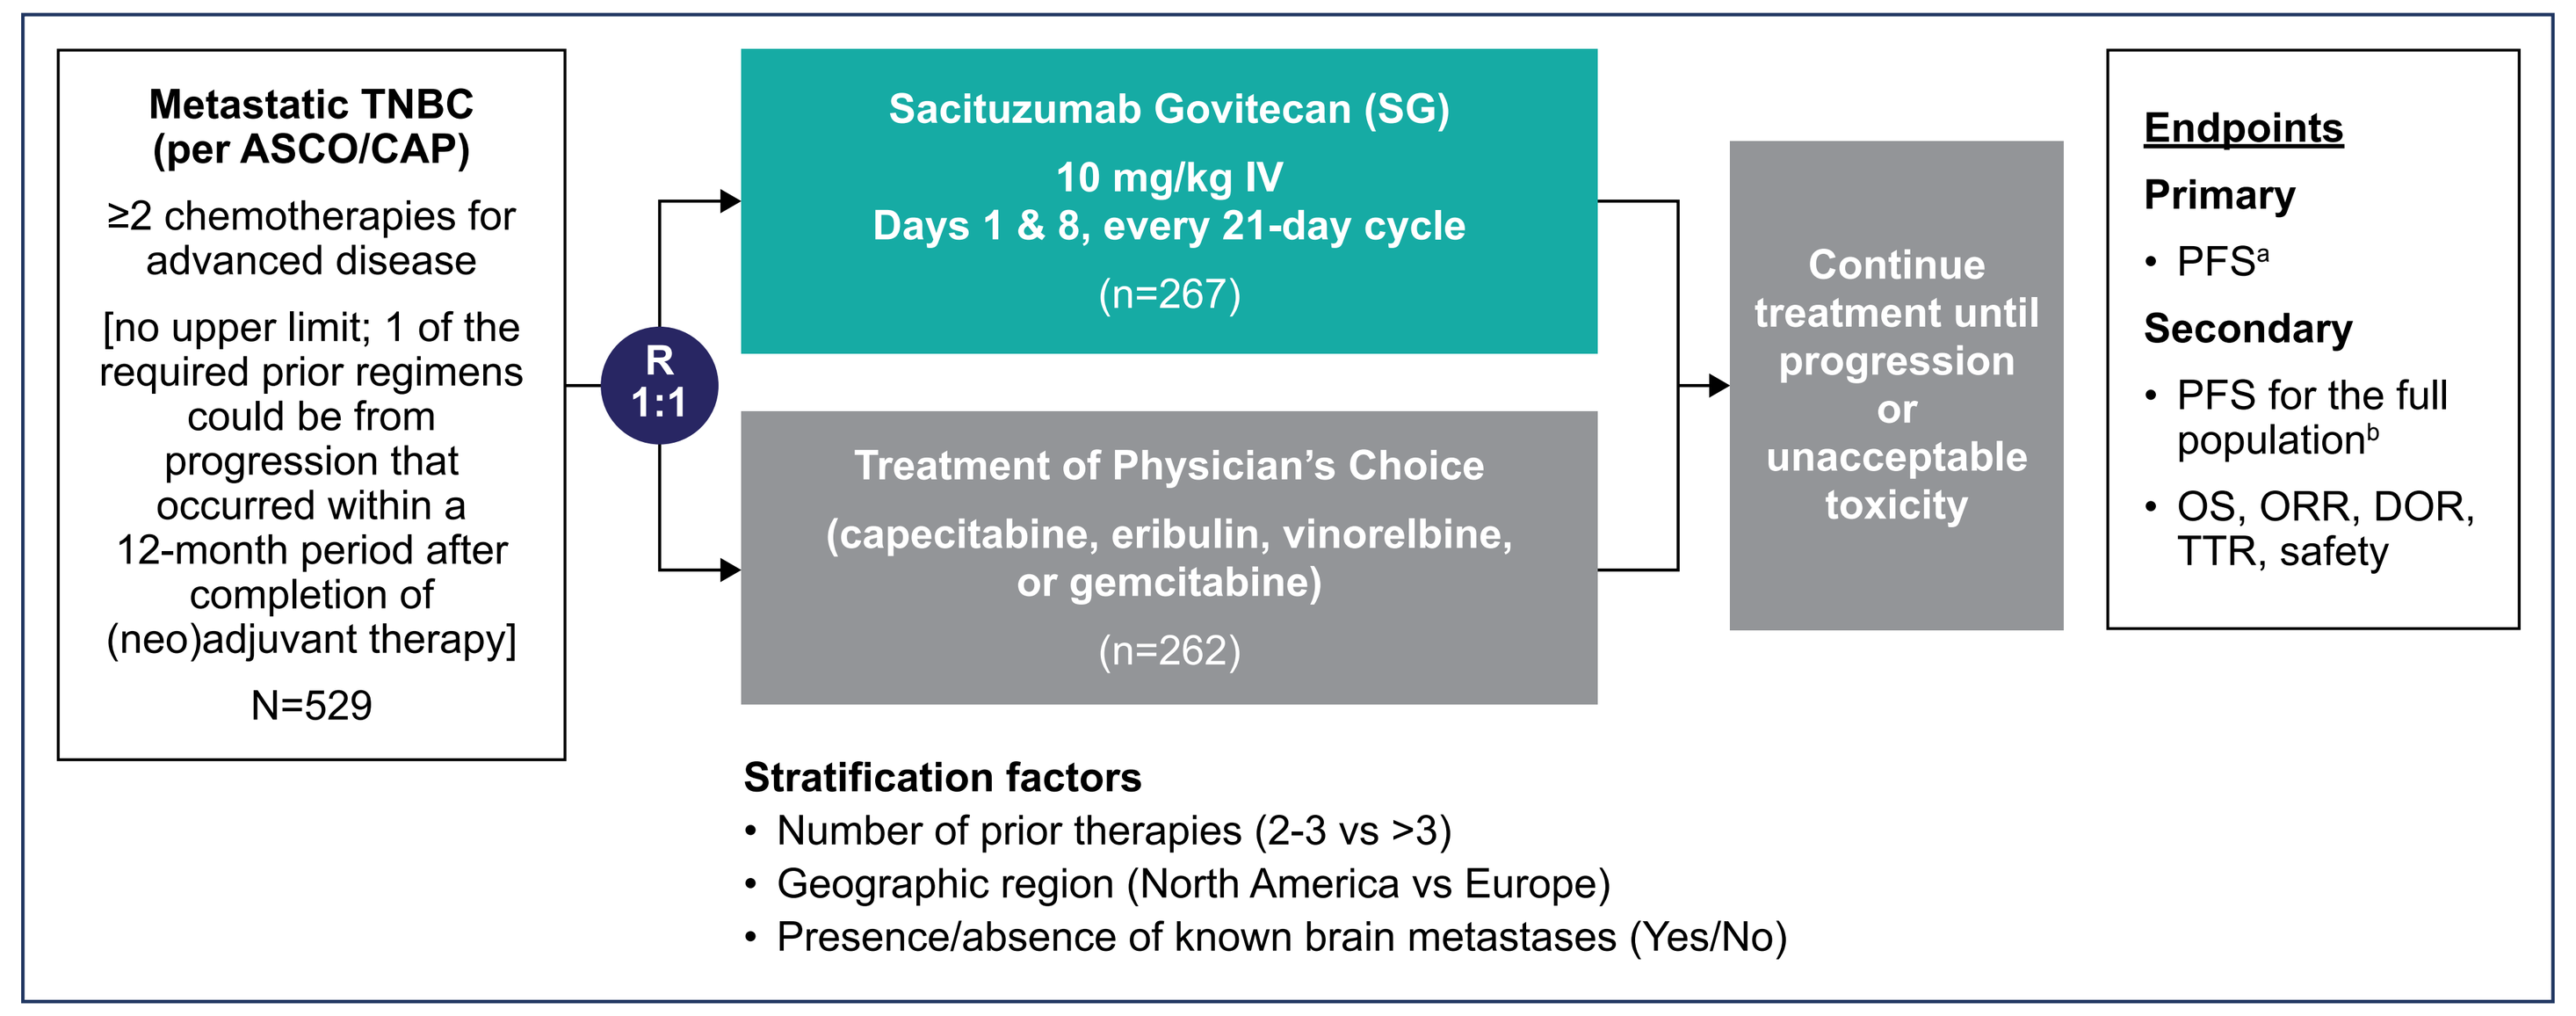

Metastatic TNBC (per ASCO/CAP)
≥2 chemotherapies for advanced disease
[no upper limit; 1 of the required prior regimens could be from progression that occurred within a 12-month period after completion of (neo)adjuvant therapy]
N=529
R 1:1
Sacituzumab Govitecan (SG)
10 mg/kg IV
Days 1 & 8, every 21-day cycle
(n=267)
Treatment of Physician's Choice
(capecitabine, eribulin, vinorelbine, or gemcitabine)
(n=262)
Continue treatment until progression or unacceptable toxicity
Endpoints
Primary
• PFS[a]
Secondary
• PFS for the full population[b]
• OS, ORR, DOR, TTR, safety
Stratification factors
• Number of prior therapies (2-3 vs >3)
• Geographic region (North America vs Europe)
• Presence/absence of known brain metastases (Yes/No)

Adapted from *N Engl J Med*. Bardia A, Hurvitz SA, Tolaney SM, et al. Sacituzumab govitecan in metastatic triple-negative breast cancer. Vol. 384, pp 1529-1541. Copyright ©2021 Massachusetts Medical Society. Reused with permission from Massachusetts Medical Society.
[a]PFS measured by an independent, centralized, and blinded group of radiology experts who assessed tumor response using RECIST 1.1 criteria in patients without brain metastasis.
[b]The full population includes all randomized patients (with and without brain metastases). Baseline brain MRI only required for patients with known brain metastasis.
ASCO/CAP, American Society of Clinical Oncology/College of American Pathologists; DOR, duration of response; IV, intravenous; mTNBC, metastatic triple-negative breast cancer; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; R, randomization; RECIST 1.1, Response Evaluation Criteria in Solid Tumors, version 1.1; SG, sacituzumab govitecan; TNBC, triple-negative breast cancer; TPC, treatment of physician's choice; TTR, time to response. National Institutes of Health. https://clinicaltrials.gov/ct2/show/NCT02574455.

Results

Patients

- A total of 529 patients were enrolled in the ASCENT trial (full trial population), of which 62 (12%) were Black patients (**Table 1**)
 - Of these patients, 28 were randomized to receive SG and 34 to receive TPC
- Baseline characteristics were generally balanced across treatment arms (**Table 1**)
 - Two patients (3%; 1 each in the SG and TPC arms) were positive for *BRCA1/2* mutations
 - Six patients (18%) in the TPC arm had known brain metastases at study entry vs none in the SG arm
- Median treatment duration was 5.3 vs 1.6 months in the SG and TPC arms, respectively

Table 1. Demographics and Baseline Characteristics of ITT Population Reporting Black Race

	SG (n=28)	TPC (n=34)
Female, n (%)	28 (100)	34 (100)
Median age at study entry, y (range)	50 (35-69)	55 (32-75)
ECOG performance status, n (%)		
0	12 (43)	14 (41)
1	16 (57)	20 (59)
Known brain metastases at study entry, n (%)	0	6 (18)
TNBC at initial breast cancer diagnosis, n (%)	22 (79)	27 (79)
Number of prior chemotherapies, n (%)		
2-3	21 (75)	22 (65)
>3	7 (25)	12 (35)
Median prior systemic regimens,[a] n (range)	4 (2-8)	4 (2-10)
Previous use of checkpoint inhibitors, n (%)	11 (39)	12 (35)
Setting of prior systemic therapies, n (%)		
Adjuvant	16 (57)	17 (50)
Neoadjuvant	13 (46)	16 (47)
Metastatic	25 (89)	34 (100)
Locally advanced disease	2 (7)	0
***BRCA1/2* mutational status, n (%)**		
Negative	13 (46)	17 (50)
Positive	1 (4)	1 (3)
Unknown	14 (50)	16 (47)

[a]Prior systemic anti-cancer regimens are defined as regimens with a start and end date prior to first administration of study treatment.
BRCA, breast cancer gene; ECOG, Eastern Cooperative Oncology Group; SG, sacituzumab govitecan; TNBC, triple-negative breast cancer; TPC, treatment of physician's choice; y, years.

Results *(continued)*

Efficacy in ITT Population Reporting Black Race

- Median PFS was improved with SG vs TPC (5.4 vs 2.2 months; HR, 0.44; 95% CI, 0.24-0.80; *P*=0.008; **Figure 3**)
- Median OS was longer with SG vs TPC (13.8 vs 8.5 months; HR, 0.64; 95% CI, 0.34-1.19; *P*=0.159; **Figure 4**)
- ORR with SG vs TPC was 32% vs 6% (**Table 2**)
 - In the SG arm, 1 patient (4%) had a complete response (CR) and 8 patients (29%) had a partial response (PR)
 - In the TPC arm, 1 patient (3%) had a CR and 1 patient (3%) had a PR
- Median duration of response (DOR) was 9.2 months in the SG arm and not evaluable in the TPC arm (**Table 2**)
- Efficacy outcomes for SG in Black patients were consistent with those of SG in the full ASCENT study population[17]

Figure 3. Progression-Free Survival

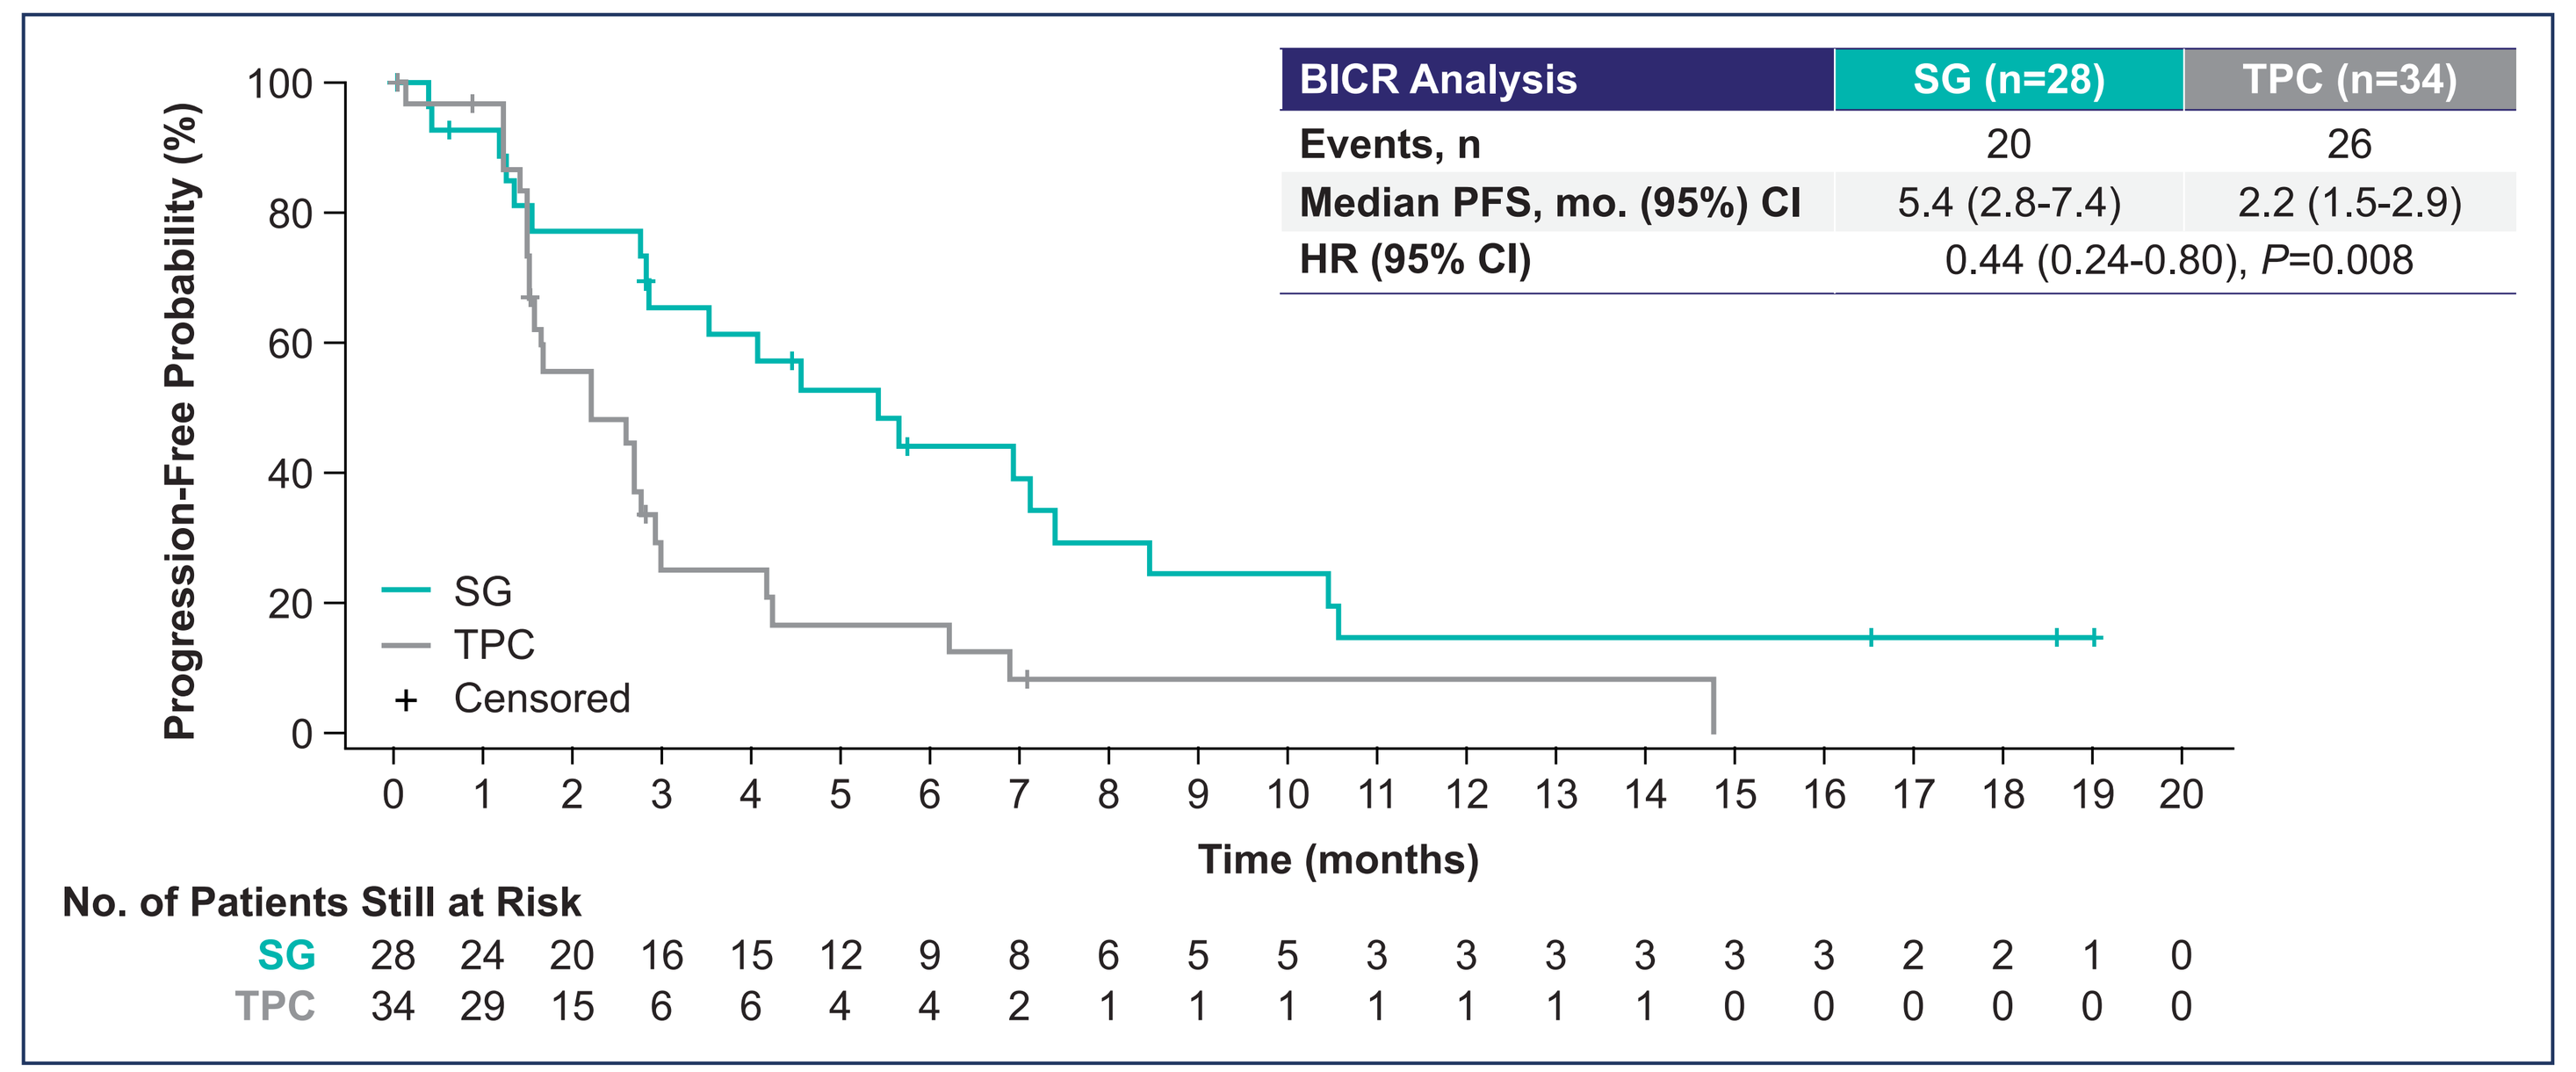

BICR Analysis | SG (n=28) | TPC (n=34)
Events, n | 20 | 26
Median PFS, mo. (95%) CI | 5.4 (2.8-7.4) | 2.2 (1.5-2.9)
HR (95% CI) | 0.44 (0.24-0.80), P=0.008
Progression-Free Probability (%)
SG
TPC
+ Censored
Time (months)
No. of Patients Still at Risk
SG 28 24 20 16 15 12 9 8 6 5 5 3 3 3 3 3 3 2 2 1 0
TPC 34 29 15 6 6 4 4 2 1 1 1 1 1 1 1 0 0 0 0 0 0

BICR, blinded independent central review; PFS, progression-free survival; SG, sacituzumab govitecan; TPC, treatment of physician's choice.

Figure 4. Overall Survival

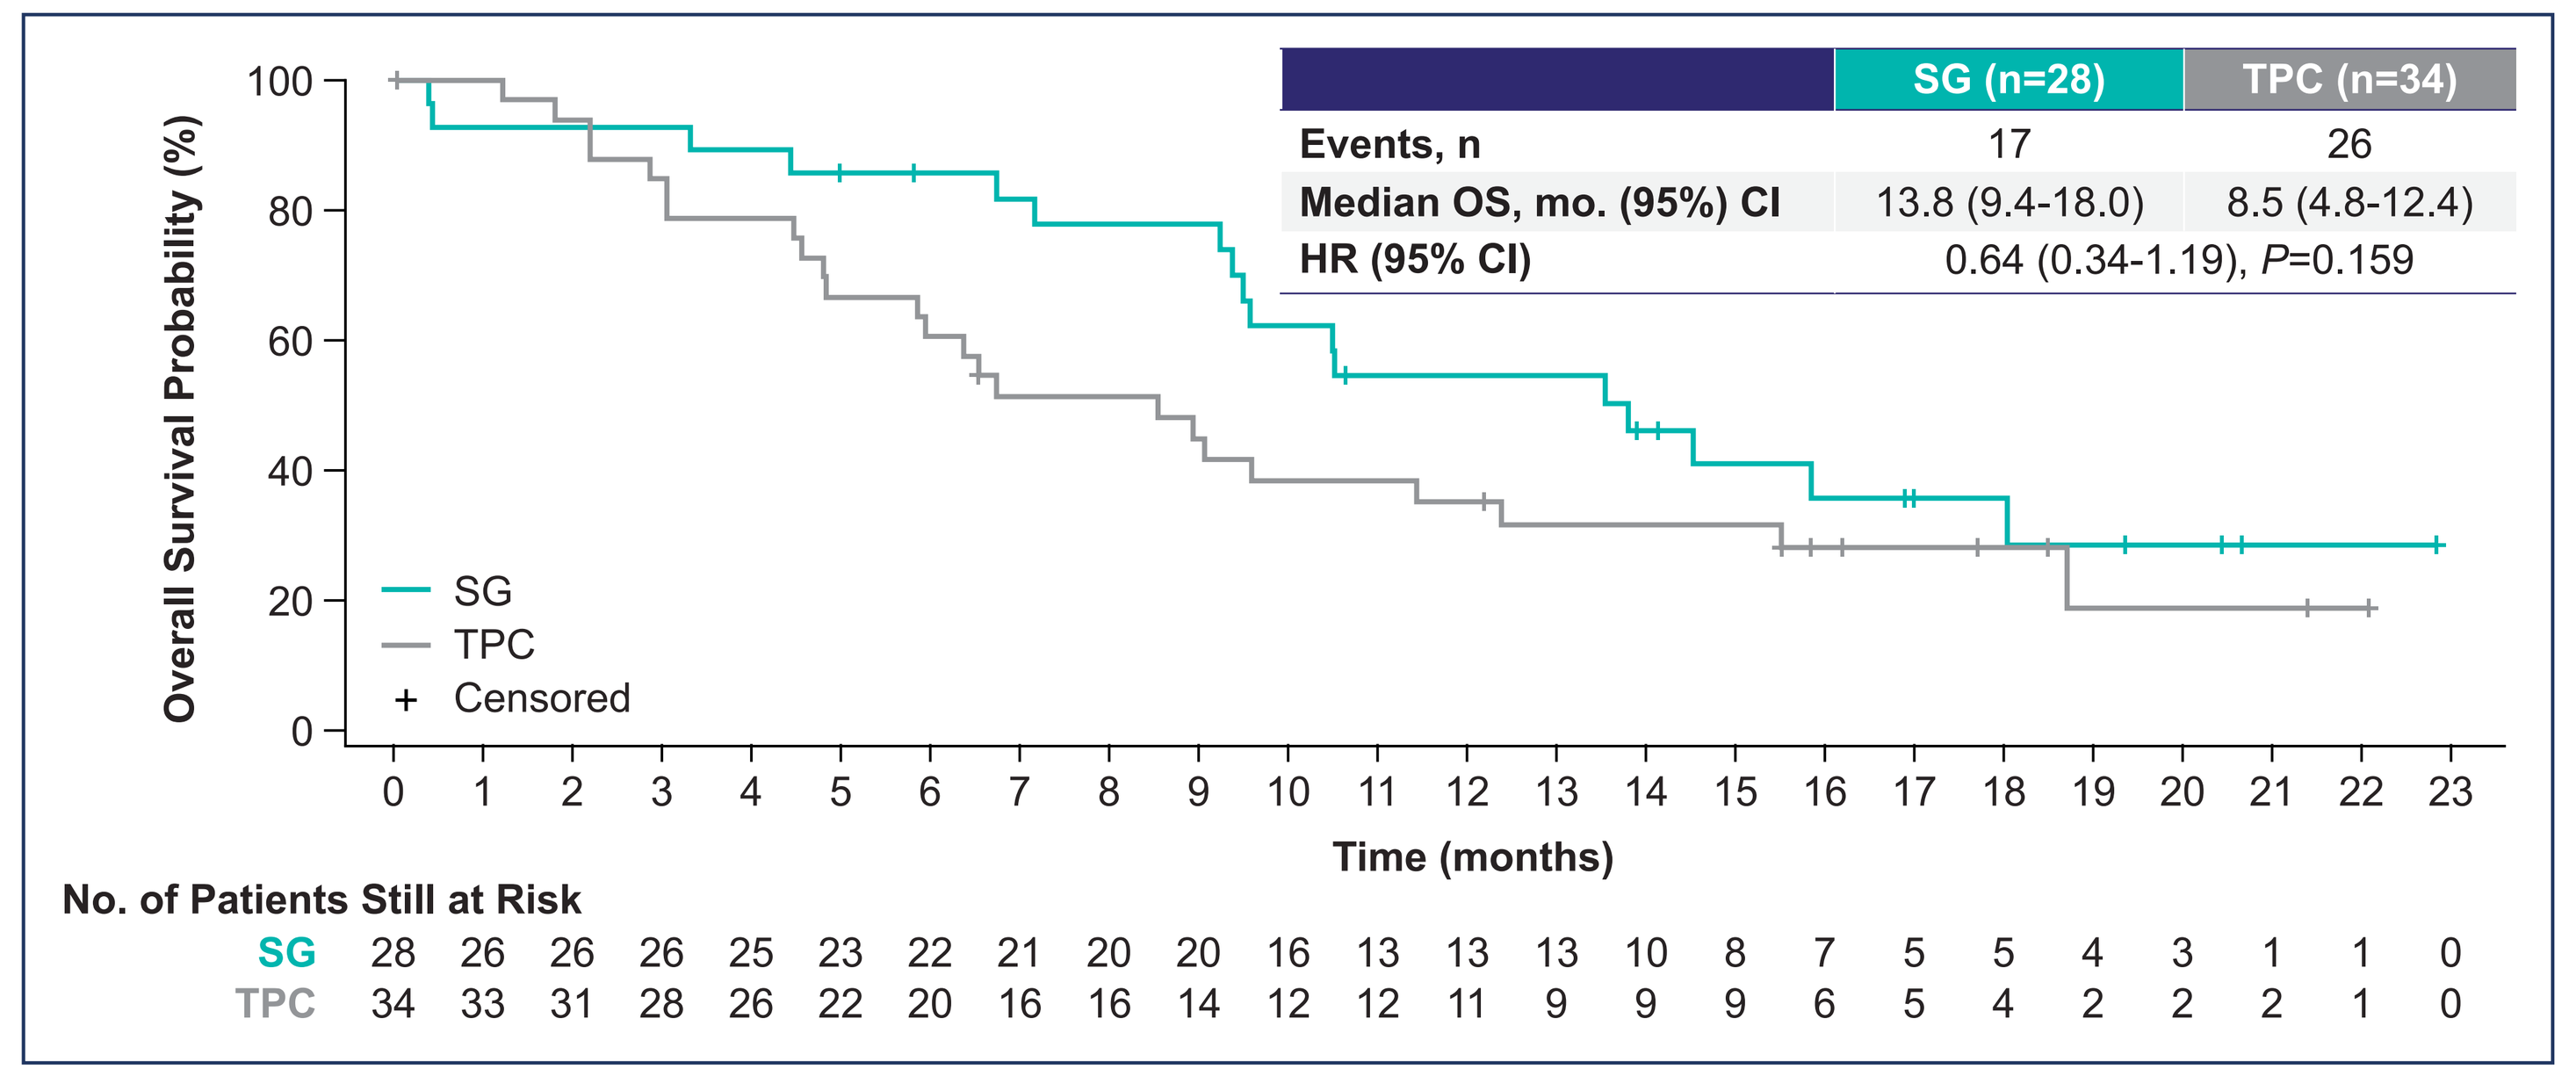

SG (n=28) | TPC (n=34)
Events, n | 17 | 26
Median OS, mo. (95%) CI | 13.8 (9.4-18.0) | 8.5 (4.8-12.4)
HR (95% CI) | 0.64 (0.34-1.19), P=0.159
Overall Survival Probability (%)
SG
TPC
+ Censored
Time (months)
No. of Patients Still at Risk
SG 28 26 26 26 25 23 22 21 20 20 16 13 13 13 10 8 7 5 5 4 3 1 1 0
TPC 34 33 31 28 26 22 20 16 16 14 12 12 11 9 9 9 6 5 4 2 2 2 1 0

OS, overall survival; SG, sacituzumab govitecan; TPC, treatment of physician's choice

Table 2. Overall Response

	SG (n=28)	TPC (n=34)
ORR, n (%)	9 (32)	2 (6)
Best overall response, n (%)		
CR	1 (4)	1 (3)
PR	8 (29)	1 (3)
SD	11 (39)	12 (35)
SD >6 months	3 (11)	3 (9)
PD	4 (14)	14 (41)
Not evaluable	4 (14)	6 (18)
CBR,[a] n (%)	12 (43)	5 (15)
Median DOR, mo. (95% CI)	9.2 (3.2-NE)	NE (2.9-NE)

[a]Complete response, partial response and stable disease ≥6 months, combined.
BOR, best overall response; CBR, clinical benefit rate; CR, complete response; DOR, duration of response; NE, not evaluable; ORR, objective response rate; PR, partial response; SD, stable disease; SG, sacituzumab govitecan; TPC, treatment of physician's choice.

Safety in ITT Population Reporting Black Race

- The most common treatment-related adverse events (TRAEs; SG vs TPC) of any grade in this subgroup (**Table 3**) were neutropenia (64% vs 61%), diarrhea (64% vs 13%), and fatigue (52% vs 39%)
- The most common TRAEs (SG vs TPC) of grade ≥3 in this subgroup (**Table 3**) were neutropenia (48% vs 42%), anemia (12% vs 6%), leukopenia (8% vs 16%), and febrile neutropenia (8% vs 3%)
 - The safety profile of SG in this subgroup was consistent with that of the full trial population[17]
- No SG-related events of severe cardiovascular toxicity, grade >1 neuropathy, or interstitial lung disease were reported in this subgroup analysis
- Dose reductions due to treatment-emergent adverse events occurred in 28% vs 35% of patients in the SG vs TPC arms
- Discontinuations due to TRAEs occurred in 0% vs 3% of patients in the SG vs TPC arms
- No treatment-related deaths occurred in either arm

Table 3. TRAEs of Any Grade (≥20%) and Grade ≥3 (≥5%)

		SG (n=25)			TPC (n=31)		
TRAE[a]		**All grade, n (%)**	**Grade 3, n (%)**	**Grade 4, n (%)**	**All grade, n (%)**	**Grade 3, n (%)**	**Grade 4, n (%)**
Hematologic	**Neutropenia[b]**	16 (64)	9 (36)	3 (12)	19 (61)	9 (29)	4 (13)
	Anemia[c]	10 (40)	3 (12)	0	9 (29)	2 (6)	0
	Leukopenia[d]	6 (24)	2 (8)	0	10 (32)	4 (13)	1 (3)
	Febrile neutropenia	2 (8)	2 (8)	0	1 (3)	0	1 (3)
Gastrointestinal	**Nausea**	11 (44)	0	1 (4)	11 (36)	0	0
	Diarrhea	16 (64)	1 (4)	0	4 (13)	0	0
	Vomiting	9 (36)	0	1 (4)	8 (26)	0	0
	Constipation	2 (8)	0	0	9 (29)	0	0
Other	**Fatigue**	13 (52)	0	0	12 (39)	3 (10)	0
	Alopecia	9 (36)	0	0	3 (10)	0	0

[a]Treatment-related adverse events have a start date on or after the date of first dose of study treatment and up to 30 days after date of last dose of study treatment. Patients may report more than one per system organ class or preferred term. At each level of summarization, a patient is counted once if he/she reported one or more adverse events. [b]Combined preferred terms of 'neutropenia' and 'neutrophil count decreased.' [c]Combined preferred terms of 'anemia', 'hemoglobin decreased', and 'red blood cell count decreased'. [d]Combined preferred terms of 'leukopenia' and 'white blood cell count decreased'.
SG, sacituzumab govitecan; TPC, treatment of physician's choice; TRAE, treatment-related adverse events.

Conclusions

- In the phase 3 ASCENT study of SG in refractory/relapsed mTNBC, 12% of patients self-reported their race as Black
- Black patients treated with SG in the 2L+ mTNBC setting derived a similar clinical benefit from SG over TPC, consistent with that of the full trial population[17]
 - Median PFS of 5.4 vs 2.2 months (HR, 0.44; 95% CI, 0.24-0.80; *P*=0.008)
 - Median OS of 13.8 vs 8.5 months (HR, 0.64; 95% CI, 0.34-1.19; *P*=0.159)
 - ORR of 32% vs 6%
- The safety profile of SG in Black patients from ASCENT was manageable, and consistent with that of the full trial population[17]
- SG should be considered a treatment option for Black patients with mTNBC who have received ≥2 prior chemotherapies, with at least 1 in the metastatic setting

References

1. Berrocal JK, et al. *Am J Hematol* Oncol. 2017;13:16-19.
2. Kohler BA, et al. *J Natl Cancer Inst*. 2015;107:djv048.
3. Plasilova ML, et al. *Medicine*. 2016;95:35(e4614).
4. Brufsky A, et al. *Breast Cancer Res Treat*. 2012;133:1067-1075.
5. Pivot X, et al. *Ann Oncol*. 2016;27:1525-1531.
6. Perez EA, et al. *Breast Cancer Res Treat*. 2010;121:261-271.
7. Park IH, et al. *Cancer Res Treat*. 2019;51:43-52.
8. Dietze EC, et al. *Nat Rev Cancer*. 2015;15:248-254.
9. Cho B, et al. *JAMA Oncol*. 2021;7:1016-1023.
10. DeSantis CE, et al. *CA Cancer J Clin*. 2019;69:438-451.
11. Goldenberg DM, et al. *Expert Opin Biol Ther*. 2020;20:871-885.
12. Nagayama A, et al. *Ther Adv Med Oncol*. 2020;12:1758835920915980.
13. Goldenberg DM, et al. *Oncotarget*. 2015;6:22496-224512.
14. Cardillo TM, et al. *Bioconjugate Chem*. 2015;26:919-931.
15. Govindan SV, et al. *Mol Cancer Ther*. 2013;12:968-978.
16. TRODELVY™ (sacituzumab govitecan-hziy). Prescribing Information. Immunomedics, Inc.; April 2021.
17. Bardia A, et al. *N Engl J Med*. 2021;384:1529-1541.
18. Food and Drug Administration. FDA grants regular approval to sacituzumab govitecan for triple-negative breast cancer. https://www.fda.gov/drugs/resources-information-approved-drugs/fda-grants-regular-approval-sacituzumab-govitecan-triple-negative-breast-cancer. Accessed September 28, 2021.

Acknowledgments

- We thank the patients and their caregivers for helping us realize the possibilities of this research
- We thank the dedicated clinical trial investigators and their devoted team members for participating in the ASCENT trial
- This study is sponsored by Gilead Sciences, Inc.
- Editorial support was provided by Team9Science and funded by Gilead Sciences, Inc.

Disclosures

- Lisa A. Carey reports research funding from Sanofi, Novartis, Genentech/Roche, and GSK; spouse serves on the board of Falcon Therapeutics

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SABCS® and the author of this poster.

Presented at the San Antonio Breast Cancer Symposium – December 7-10, 2021

This presentation is the intellectual property of the author/presenter. Contact them at Lisa_Carey@med.unc.edu for permission to reprint and/or distribute.

©2021 Gilead Sciences, Inc. All rights reserved.